ASX Announcement

25 October 2019

ANP 2nd Neuromuscular Drug Development Summit Poster Presentation

Antisense Therapeutics Ltd (ASX:ANP) is pleased to advise that Dr George Tachas, ANP's Director of Drug Discovery and Patents has today given his poster presentation at the 2nd Neuromuscular Drug Development Summit (NMDS) in Boston, MA, USA.

The poster presented entitled "Preliminary results from a CD49d antisense drug ATL1102 6 month Phase II trial in non-ambulant patients with Duchenne's Muscular Dystrophy" follows this announcement.

As previously advised (17 October 2019), the NMDS is the first occasion for ANP to showcase its ATL1102 DMD project and positive preliminary trial results to a biopharmaceutical community focused on DMD and other neuromuscular diseases.

For more information please contact:

Antisense Therapeutics
Mark Diamond
Managing Director
+61 (0)3 9827 8999
www.antisense.com.au

Investment Enquiries
Gennadi Koutchin
XEC Partners
gkoutchin@xecpartners.com.au
1300 932 037

About Antisense Therapeutics Limited (ASX: ANP) is an Australian publicly listed biopharmaceutical company, developing and commercialising antisense pharmaceuticals for large unmet markets. The products are in-licensed from Ionis Pharmaceuticals Inc. (NASDAQ:IONS), world leaders in antisense drug development and commercialisation. ATL1102 (injection) has successfully completed a Phase II efficacy and safety trial, significantly reducing the number of brain lesions in patients with relapsing-remitting multiple sclerosis (RR-MS). ATL1103 drug designed to block GHr production successfully reduced blood IGF-I levels in Phase II clinical trials in patients with the growth disorder acromegaly. The Company is conducting a Phase II clinical trial of ATL1102 in DMD patients at the Royal Childrens Hospital, Melbourne.

About ATL1102 ATL1102 is an antisense inhibitor of CD49d, a subunit of VLA-4 (Very Late Antigen-4). Antisense inhibition of VLA-4 expression has demonstrated activity in a number of animal models of inflammatory disease including asthma and MS with the MS animal data having been published in a peer reviewed scientific journal. ATL1102 was shown to be highly effective in reducing MS lesions in a Phase IIa clinical trial in RR-MS patients. The ATL1102 Phase IIa clinical data has been published in the medical Journal ***Neurology*** (Limmroth, V. et al Neurology, 2014; 83(20): 1780-1788).

About ATL1102 DMD Trial The Company is undertaking a clinical trial of ATL1102 in patients with Duchenne Muscular Dystrophy. The open label six-month dosing trial of ATL1102 in nine non-ambulant patients with DMD aged between 10 and 18 years is being conducted at the neuromuscular centre of the Royal Children's Hospital (RCH) which operates the largest clinic in the southern hemisphere treating children with DMD. The primary endpoints of the trial relate to the safety and tolerability of ATL1102. The efficacy of ATL1102 will also be assessed in terms of its effects on disease processes and progression (e.g. the upper limb strength and function of the boys). Further details on the trial are available here on the Australia and New Zealand Clinical Trials Registry.

6 WALLACE AVENUE, TOORAK VIC 3142 AUSTRALIA TEL . +61 (3) 9827 8999 FAX +61 (3) 9827 1166 WEB WWW.ANTISENSE.COM.AU
ANTISENSE THERAPEUTICS LIMITED ABN 41 095 060 745

About DMD Duchenne Muscular Dystrophy (DMD) is an X-linked disease that affects 1 in 3600 to 6000 live male births (Bushby *et al*, 2010). DMD occurs as a result of mutations in the dystrophin gene which causes a substantial reduction in or absence of the dystrophin protein. Children with DMD have dystrophin deficient muscles and are susceptible to contraction induced injury to muscle that triggers the immune system which exacerbates muscle damage as summarized in a publication co-authored by the Director of the FDA CDER (Rosenberg et al, 2015). Ongoing deterioration in muscle strength affects lower limbs leading to impaired mobility, and also affects upper limbs, leading to further loss of function and self-care ability. The need for wheelchair use can occur in early teenage years for patients on corticosteroids with a mean age of 13, with respiratory, cardiac, cognitive dysfunction also emerging. Patients with a greater number of immune T cells expressing high levels of CD49d have more severe and progressive disease and are wheelchair bound by the age of 10 despite being on corticosteroid treatment (Pinto Mariz et al, 2015). With no intervention, the mean age of life is approximately 19 years. The management of the inflammation associated with DMD is currently addressed via the use of corticosteroids, however they are acknowledged as providing insufficient efficacy and are associated with significant side effects. As a consequence, there is an acknowledged high need for new therapeutic approaches for the treatment of inflammation associated with DMD.

Rosenberg AS, Puig M, Nagaraju K, *et al*. Immune-mediated pathology in Duchenne muscular dystrophy. *Sci Transl Med* 2015, 7: 299rv4.

Busby et al for the DMD Care Consideration Working Group/ *Diagnosis and management of Duchenne muscular dystrophy, part 1* Lancet Neurol. **2010** Jan;9(1):77-93 *and part 2* Lancet Neurol. **2010** Feb;9(2):177-89 *.*

Pinto-Mariz F, Carvalho LR, Araújo AQC, *et al*. CD49d is a disease progression biomarker and a potential target for immunotherapy in Duchenne muscular dystrophy. *Skeletal Muscle* 2015, 5: 45-55

ANTISENSE THERAPEUTICS LIMITED ABN 41 095 060 745

Preliminary results from a CD49d antisense drug ATL1102 6 month Phase II trial in non-ambulant patients with Duchenne's Muscular Dystrophy

G Tachas[1], N Desem[1], Monique M. Ryan[2], and Ian Woodcock[2.]

[1]Antisense Therapeutics Limited, Melbourne, Victoria, Australia. www.antisense.com.au

[2] Department of Neurology, Royal Children's Hospital, Melbourne Australia

Introduction

The safety and activity of ATL1102, an antisense drug to the RNA for CD49d, the alpha subunit of VLA-4 [1], is being evaluated in a 24 week dosing study in non-ambulant patients with Duchenne's Muscular Dystrophy (DMD). DMD is an X linked inherited disease caused by mutations in the dystrophin gene resulting in muscle breakdown from contraction, and immune mediated inflammatory damage, leading to loss of ambulation, upper limb function and respiratory and cardiac failure. DMD immune mediated inflammatory damage is currently treated with corticosteroids but there is a need for safer and more effective treatments. Patients with DMD who have a greater number of immune T cells with high levels of CD49d expression have more severe and rapid disease progression despite corticosteroid use[2]. ATL1102 was previously shown to reduce disease activity in a Phase II multiple sclerosis study[1].

[2.] Pinto-Mariz et al (2015). Skeletal Muscle; 5: 45-55

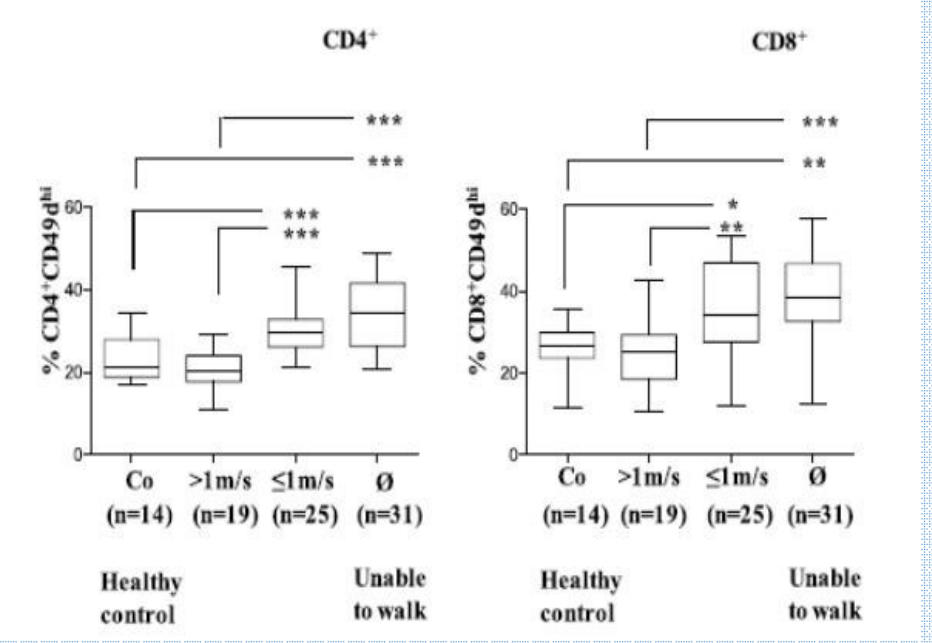

CD4+
CD8+
% CD4+CD49d hi
% CD8+CD49d hi

**
*
0
20
40
60
Co
>1m/s
≤1m/s
Ø
(n=14)
(n=19)
(n=25)
(n=31)
Healthy control
Unable to walk

- Healthy Controls (Co) and DMD patients who walked >1m/s had ~ 20% CD4+CD49d+ hi and ~25% CD8+ CD49d+ hi T-cells
- DMD patients who walk <1m/s and non ambulant patients had ~35% CD4+CD49d+ hi and ~40% CD8+ CD49d+ hi T-cells

Methods

In a single centre, open label Phase II study, nine adolescent non-ambulatory DMD male patients (10-18 years) are being treated with 25mg of ATL1102 subcutaneously injected once weekly for 24 weeks and monitored for a further 8 weeks. The primary study endpoints relate to safety and tolerability with secondary endpoints to assess the drug activity via effects on white blood cells and on disease progression via muscle strength and functional analysis (ACTRN12618000970246)[3]. A review was undertaken of the preliminary data from the first 6 patients who had completed their 24 weeks of dosing.

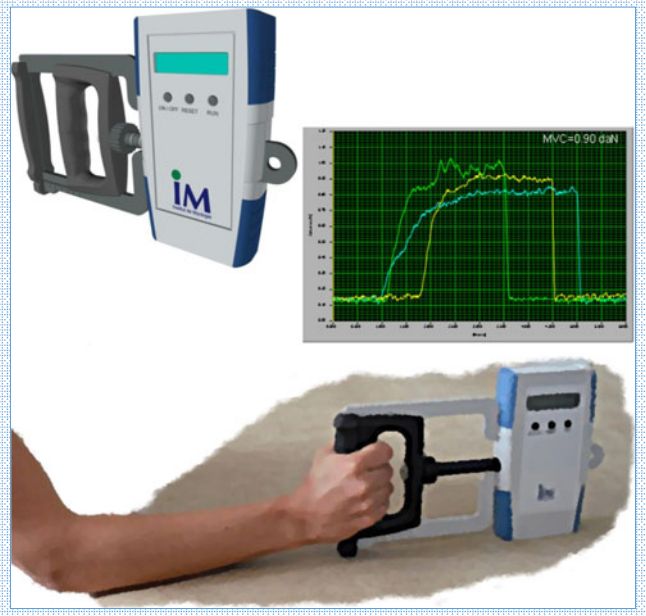

MVC=0.90 daN

Myo-Grip

Myo-Pinch

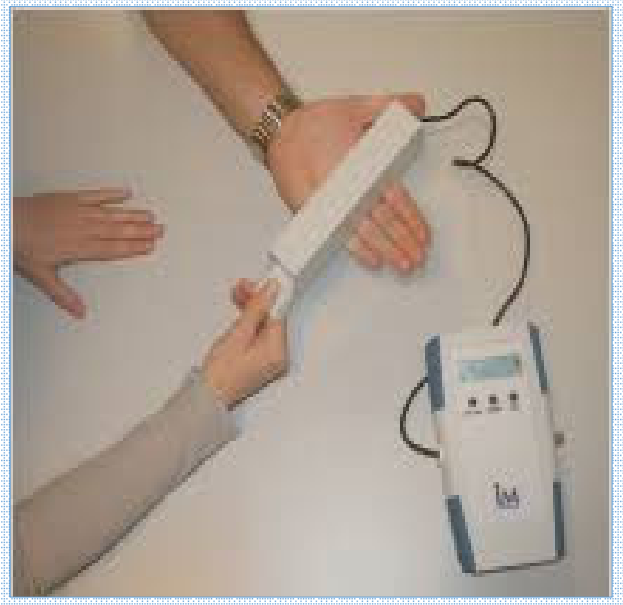

www.institut-myologie.org/

Results

The preliminary data review suggested a positive drug effect of ATL1102 at the dose tested both at immunomodulatory (i.e. effects on relevant immune cells) and disease progression (i.e. effects on muscle strength) levels. With respect to the safety related trial data, no Serious Adverse Events (SAE's) had been reported to date. The Data Safety Monitoring Board had evaluated the safety data at that time and recommended continuation of the trial with no safety concerns.

The number of immune T cells expressing CD49d, were trending downward during the 24 week treatment phase while returning to around starting levels post dosing.

As an indicator of ATL1102's suggestive effects on disease progression the patients pinch and grip strength as assessed by the Myo-Pinch and Myo-Grip device increased from baseline by a mean +0.1 kg 95% CI [-0.23, 0.34] and +0.3 kg 95% CI [-0.38, 1.08] respectively.

This is compared to losses in muscle pinch and grip strength of -0.38kg 95% CI [-0.53, -0.22] and -0.5 kg [-1.01, 0.002] respectively reported in a study in 9 non-ambulant DMD patients on corticosteroids assessed at 6 months[4].

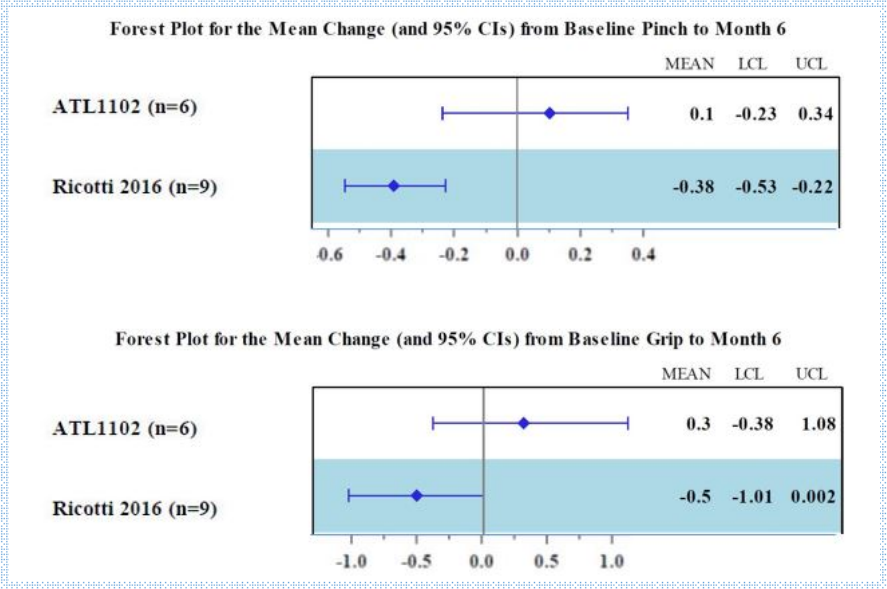

Forest Plot for the Mean Change (and 95% CIs) from Baseline Pinch to Month 6
MEAN LCL UCL
ATL1102 (n=6) 0.1 -0.23 0.34
Ricotti 2016 (n=9) -0.38 -0.53 -0.22
-0.6 -0.4 -0.2 0.0 0.2 0.4
Forest Plot for the Mean Change (and 95% CIs) from Baseline Grip to Month 6
MEAN LCL UCL
ATL1102 (n=6) 0.3 -0.38 1.08
Ricotti 2016 (n=9) -0.5 -1.01 0.002
-1.0 -0.5 0.0 0.5 1.0

https://www.antisense.com.au/index.php/asx-announcements/

Conclusions

In an ongoing study of ATL1102 in patients with DMD treated for 24 weeks, no SAE's have been reported while preliminary trial data from the first 6 patients having completed their dosing is suggestive of a drug effect of ATL1102 on the CD49d+ T cell numbers and the upper limb muscle grip and pinch strength.

Bibliography

1. Limmroth et al (2014). Neurology, 11:83 (20) 1780-8
2. Pinto Mariz et al (2015). Skeletal Muscle; 5: 45-55.
3. https://www.anzctr.org.au/Trial/Registration/TrialReview.aspx?id=374611&isReview=true
4. Ricotti et al (2016). PLos One, 11(9) e0162542

Acknowledgements

The Authors wish to thank everyone involved in the clinical trial with ATL1102 in DMD (ACTRN12618000970246)[3]